DAILY TREATMENT WITH SILDENAFIL REVERSES ENDOTHELIAL DYSFUNCTION AND OXIDATIVE STRESS IN AN ANIMAL MODEL OF METABOLIC SYNDROME

Behr-Roussel D.[1], Oudot A.[1], Compagnie S.[1], Gorny D.[1], Le Coz O.[1], Bernabé J.[1], Wayman C.[2], Alexandre L.[1], Giuliano F.[1, 3]

[1] Pelvipharm, Domaine CNRS, 1 avenue de la terrasse, Bâtiment 5, F-91190 Gif-sur-Yvette, France – www.pelvipharm.com

[2] Pfizer Global Research and Development, Ramsgate Rd, Sandwich, Kent CT13 9NJ, UK

[3] AP-HP Raymond Poincaré hospital, Department of Neurological rehabilitation, 104 Bd R. Poincaré, F-92380 Garches, France - giuliano@cyber-sante.org

OBJECTIVES

- **Fructose consumption might be a contributing factor to the development of obesity and the accompanying cardiovascular disorders (hypertension, …) seen in the metabolic syndrome**

 Bray et al., Am J Clin Nutr (2004); Elliott et al., Am J Clin Nutr (2002); Hallfrisch, FASEB J (1990)
- **Patients with metabolic syndrome exhibit generalized endothelial dysfunction**

 Baron et al., Am J Cardiol (1999); Anderson et al., J Am Coll Cardiol (1995)
- **Daily treatment with PDE-5 inhibitors has beneficial effects on endothelial function in diabetic men**

 DeSouza et al., Diabetes Care (2002); Rosano et al., Eur Urol. (2005)

- **We aimed to determine whether chronic sildenafil in an experimental model of metabolic syndrome, the fructose-fed rat, could potentiate systemic endothelium-dependent relaxations by upregulating the NOS/cGMP pathway**
- **Moreover, since oxidative stress has been suggested to contribute to insulin resistance and associated endothelial dysfunction, we sought to determine the effects of chronic sildenafil on a potentially relevant biomarker of endothelial dysfunction, urinary 8-isoprostanes (IPT) content, a direct marker of non-enzymatic *in vivo* lipid peroxydation and the most reliable and clinically relevant marker of oxidative stress available to date.**

MATERIALS & METHODS

Experimental animals

Wistar rats (n=10-14 per group) were fed a standard chow (CONT) or a 60% fructose/5% fat (% by weight)-enriched diet for 8 weeks (FFR). From week 5 through 8, sildenafil was administered twice a day (sc, 20 mg/kg, FFR+SIL), thus reaching clinically relevant plasma concentrations *circa* 20 nM unbound known to give efficacy in man (*Pfizer Inc., data on file*), then a 1-week wash-out period from sildenafil was observed.

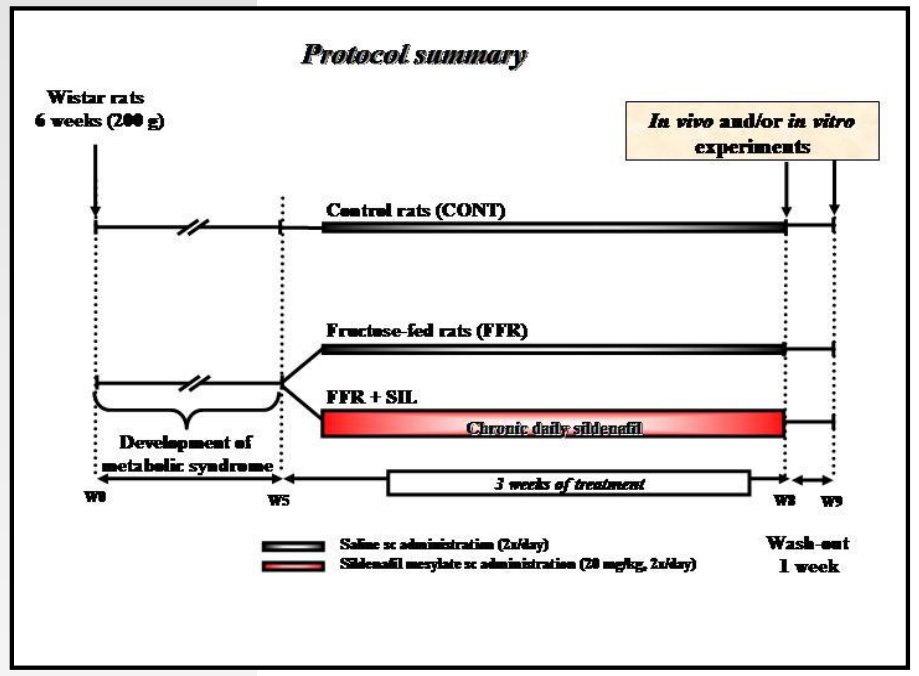
Protocol summary
Wistar rats
6 weeks (200 g)
In vivo and/or in vitro experiments
Control rats (CONT)
Fructose-fed rats (FFR)
FFR + SIL
Chronic daily sildenafil
Development of metabolic syndrome
3 weeks of treatment
W0
W5
W8
W9
Saline sc administration (2x/day)
Sildenafil mesylate sc administration (20 mg/kg, 2x/day)
Wash-out
1 week

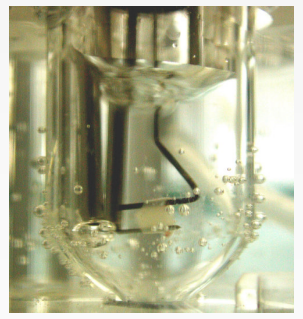

In vitro evaluation of endothelial function

Isometric tension studies were performed on isolated aortic and superior mesenteric arterial (SMA) rings precontracted with phenylephrine to build concentration-response curves (CRC) to endothelium-dependent (ACh and A23 187) and -independent (SNP) relaxants in presence of indomethacin. Vascular cGMP content, urinary excretion of nitrates and nitrites (NOx) and 8-isoprostanes (IPT), and plasma levels of IL-6 and TNF-α were also evaluated.

RESULTS

- **No influence of the fructose-enriched diet on the animal body weight, blood glucose levels, or basal mean arterial pressure (MAP)**

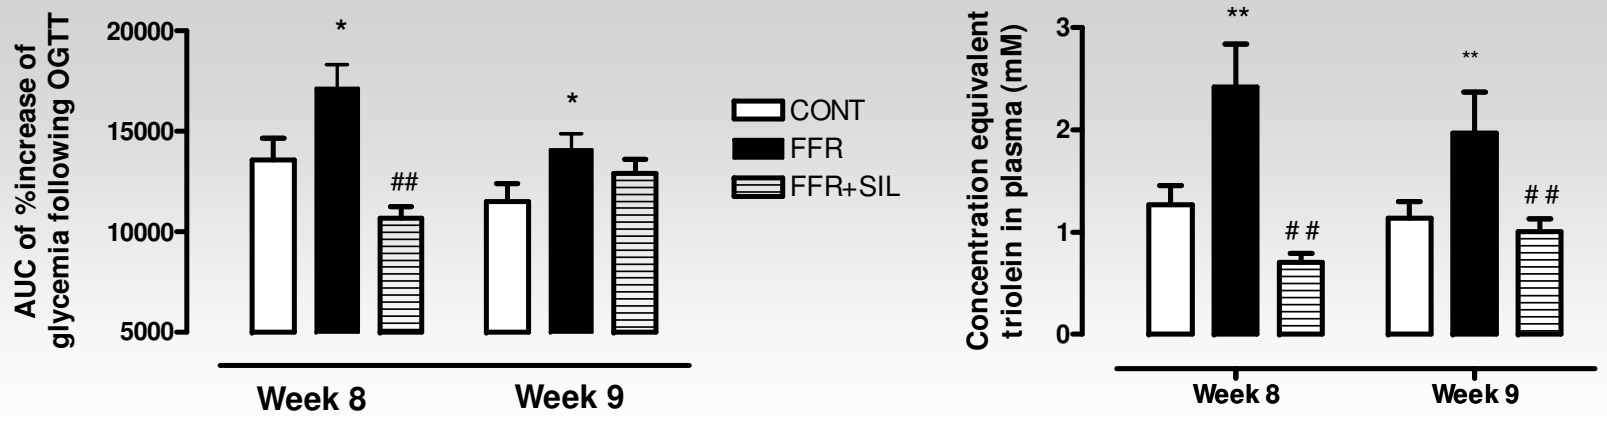
AUC of %increase of glycemia following OGTT
20000
15000
10000
5000
*
##
*
Week 8
Week 9
CONT
FFR
FFR+SIL
Concentration equivalent triolein in plasma (mM)
3
2
1
0
**
##
**
##
Week 8
Week 9

$^{*}p<0.05$, $^{**}p<0.01$ vs CONT, One-way ANOVA; $^{\#\#}p<0.01$ vs FFR, One-Way ANOVA

- **Impaired glucose tolerance in FFR corrected by a chronic treatment with sildenafil**
- **Chronic sildenafil treatment significantly countered the pronounced hypertriglyceridemia secondary to the fructose feeding in FFR and this effect was maintained even after the one-week wash-out period**

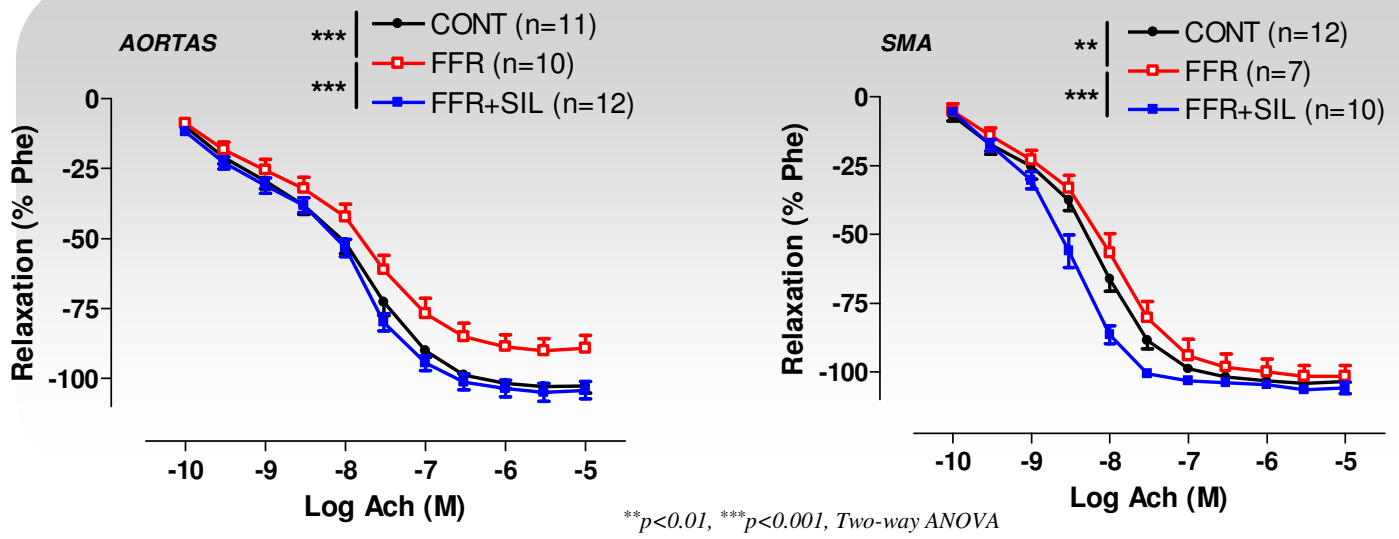
AORTAS

CONT (n=11)
FFR (n=10)
FFR+SIL (n=12)
Relaxation (% Phe)
0
-25
-50
-75
-100
-10
-9
-8
-7
-6
-5
Log Ach (M)
SMA
**

CONT (n=12)
FFR (n=7)
FFR+SIL (n=10)
Relaxation (% Phe)
Log Ach (M)

$^{**}p<0.01$, $^{***}p<0.001$, Two-way ANOVA

- **Significantly reduced endothelial relaxation responses to Ach (and A23187) in FFR in presence of indomethacin in both aortas and SMA**
- **Restoration of endothelial relaxations induced by Ach (and A23187) in FFR treated chronically with sildenafil in both aortas and SMA**
- **Enhanced compensatory endothelium-independent relaxations to SNP in FFR were not modified by sildenafil treatment**

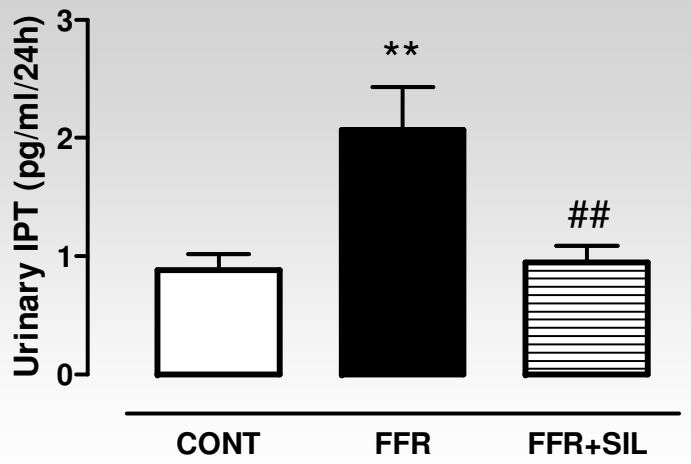
Urinary IPT (pg/ml/24h)
3
2
1
0
**
##
CONT
FFR
FFR+SIL

$^{**}p<0.01$ vs CONT, $^{\#\#}p<0.01$ vs FFR, One-way ANOVA

- **Neither the fructose diet nor the sildenafil treatment modified significantly :**
 - **Tissue basal cGMP and ET-1 content in homogenates of aortas and SMA**
 - **Urinary NOx excretion levels after one-week of wash-out from chronic sildenafil treatment**
 - **IL-6 and TNF-α plasma levels**
- **Chronic treatment with sildenafil is able to restore normal levels of urinary IPT, even one week after cessation of the treatment**

CONCLUSIONS

- **Effect of chronic sildenafil treatment on physiological parameters relevant to the metabolic syndrome : Correction of the enhanced response to glucose overload, as well as the hypertriglyceridemia induced by fructose feeding**
- **Effect of chronic sildenafil treatment on *in vitro* vascular reactivity : Restoration of normal endothelium-dependent relaxations to Ach and A23 187 in aortas and mesenteric arteries**
- **Effect of chronic sildenafil treatment on urinary 8-isoprostanes excretion : Normalization of the biological marker for oxidative stress**

The beneficial effects of daily sildenafil on vascular endothelial dysfunction gives additional insight to the possible mechanism of action of sildenafil in cardiovascular disorders related to the metabolic syndrome that could be explored in future clinical trials. Moreover, this study provides preclinical support for the predictive value of endothelial reactivity and associated biological marker of oxidative stress such as urinary IPT as surrogate markers in future clinical trials addressing cardiovascular risks.

PELVI PHARM

Study supported by a grant from Pfizer